Preclinical and Clinical Proof of Concept for Metabolic Intervention in Diabetic Cardiomyopathy

POSTER #203

Riccardo Perfetti[1], Philana Rowell[1], Shoshana Shendelman[1], Francesca Lawson[1], Ravichandran Ramasamy[2]. [1]Applied Therapeutics Inc., New York, NY; [2]NYU Langone Medical Center, New York, NY

Introduction

Diabetic Cardiomyopathy (DbCM) affects 18-24% of people with type 2 diabetes (T2D)[1,2] and currently lacks any approved therapies. Hyperactivation of the polyol pathway is a key underlying mechanism in DbCM and other diabetic complications. In hyperglycemic and ischemic conditions, polyol pathway activation causes intracellular sorbitol accumulation leading to osmotic stress, cell death and diabetic complications.[3-5] Aldose Reductase (AR), the rate-controlling enzyme in the polyol pathway, catalyzes NADPH-dependent reduction of glucose to sorbitol. Previous AR inhibitors (ARIs) failed due to safety or lack of efficacy. AT-001 is a novel oral ARI with optimized specificity and affinity for AR.

Objectives

To assess cardioprotective activity of AT-001 in an animal model† of DbCM; to determine Pharmacokinetic (PK), pharmacodynamic (PD), safety, and tolerability profile of AT-001; and to confirm target engagement in a phase 1/2 clinical study.

Methodology

Preclinical: Transgenic mice expressing human levels of AR (Tg hAR) were rendered diabetic with streptozotocin (55 mg/kg for 5 days). Mice were treated with AT-001 40 mg/kg (n=6) or vehicle (n=6) for 3 days prior to cardiac ischemia reperfusion (IR) injury induced by ligation and reperfusion of the left anterior descending coronary artery. Control non-DM Tg hAR mice (non-DM, n=9) were also subjected to IR injury. Mice were euthanized after 48 hours recovery, and infarct area and fractional shortening were assessed.

Clinical: Phase 1/2 study comprised of single oral ascending doses (SAD) of 5, 10, 20 and 40 mg/kg AT-001 or placebo once daily (QD) and multiple ascending doses (MAD) of 5, 20, 40 mg/kg AT-001 QD or 20 mg/kg twice daily (BID) or placebo for 7 days. Subjects with T2D, age 18-75 and HbA1c 5.0-8.5% were allocated to each dose cohort (n=8 AT-001, n=2 placebo).

Preclinical Study Results

At 48 hours after reperfusion:

FIGURE 1

Mean Infarct Area

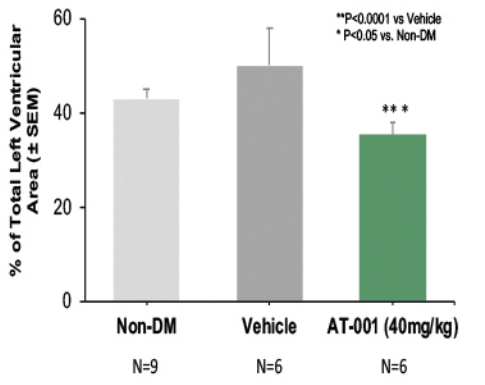
**P<0.0001 vs Vehicle
* P<0.05 vs. Non-DM
% of Total Left Ventricular Area (± SEM)
Non-DM
Vehicle
AT-001 (40mg/kg)
N=9
N=6
N=6

FIGURE 2

Mean Fractional Shortening

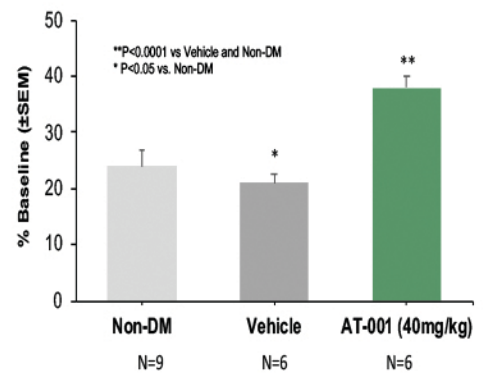
**P<0.0001 vs Vehicle and Non-DM
* P<0.05 vs. Non-DM
% Baseline (±SEM)
Non-DM
Vehicle
AT-001 (40mg/kg)
N=9
N=6
N=6

- Heart rates were not altered in different groups of mice studied.

† The ischemia reperfusion (IR) model of cardiac damage in AR transgenic mice has been validated as a relevant animal model for DbCM6. Mice naturally express very low levels of AR vs. humans, so over-expression to achieve human levels has been suggested as a more accurate model of human cardiac damage in diabetes. Cardiac damage caused by IR is greater in diabetic animals vs. non-diabetic animals, implicating AR as a key mechanism of cardiac damage during diabetes. Inhibition of AR with AT-001 reduced cardiac damage significantly in the I/R model, as measured by infarct area and fractional shortening.

Clinical Study Results

Table 1A. Demographic and Baseline Characteristics: SAD

	Placebo (N=8)	5 mg/kg (N=8)	10 mg/kg (N=8)	20 mg/kg (N=8)	40 mg/kg (N=8)	Total (N=40)
Age, years[a]	52.0 (3.5)	52.1 (2.0)	48.3 (5.9)	47.8 (4.3)	48.1 (6.5)	49.7 (4.9)
Female, n	3	6	1	4	3	17
BMI, kg/m2[a]	29.9 (2.3)	28.6 (1.4)	29.2 (2.6)	29.6 (2.6)	30.2 (2.7)	29.5 (2.3)
HbA1c, %[a]	7.3 (0.5)	7.7 (0.7)	7.2 (0.8)	7.3 (1.0)	6.3 (1.2)	7.2 (0.9)

Table 1B. Demographic and Baseline Characteristics: MAD

	Placebo (N=8)	5 mg/kg (N=8)	20 mg/kg (N=8)	40 mg/kg (N=8)	20 mg/kg BID (N=8)	Total (N=40)
Age, years[a]	52.9 (5.4)	48.9 (5.0)	51.5 (13.2)	51.4 (8.9)	61.8 (6.6)	53.3 (9.1)
Female, n	2	4	4	7	4	21
BMI, kg/m2[a]	28.4 (3.2)	29.1 (3.4)	29.2 (3.3)	29.9 (3.9)	29.2 (4.1)	29.2 (3.4)
HbA1c, %[a]	6.6 (0.9)	6.6 (1.2)	6.1 (0.7)	7.1 (0.8)	7.4 (0.7)	6.8 (1.0)

Safety

Overall, AT-001 was well tolerated in both SAD and MAD

- No treatment emergent AEs or SAEs were observed
- No treatment-related discontinuations occurred
- No abnormalities in liver or kidney function were observed

Pharmacokinetic/Pharmacodynamic

FIGURE 3. Single ascending doses

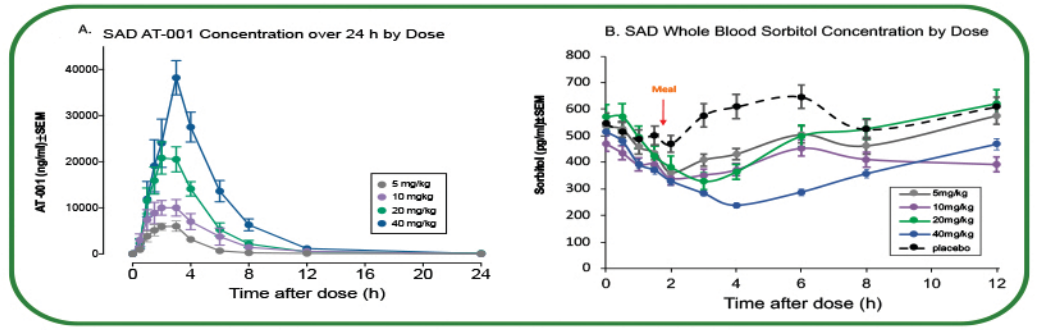
A. SAD AT-001 Concentration over 24 h by Dose
AT-001 (ng/ml)±SEM
Time after dose (h)
5 mg/kg
10 mg/kg
20 mg/kg
40 mg/kg
B. SAD Whole Blood Sorbitol Concentration by Dose
Sorbitol (pg/ml)±SEM
Meal
Time after dose (h)
5mg/kg
10mg/kg
20mg/kg
40mg/kg
placebo

Note rise in sorbitol in placebo group at ~2h (red arrow) reflects the effect of food intake (AR activation in response to post prandial glucose elevations) and it is blunted by AT-001 in patients

FIGURE 4. Multiple ascending dose

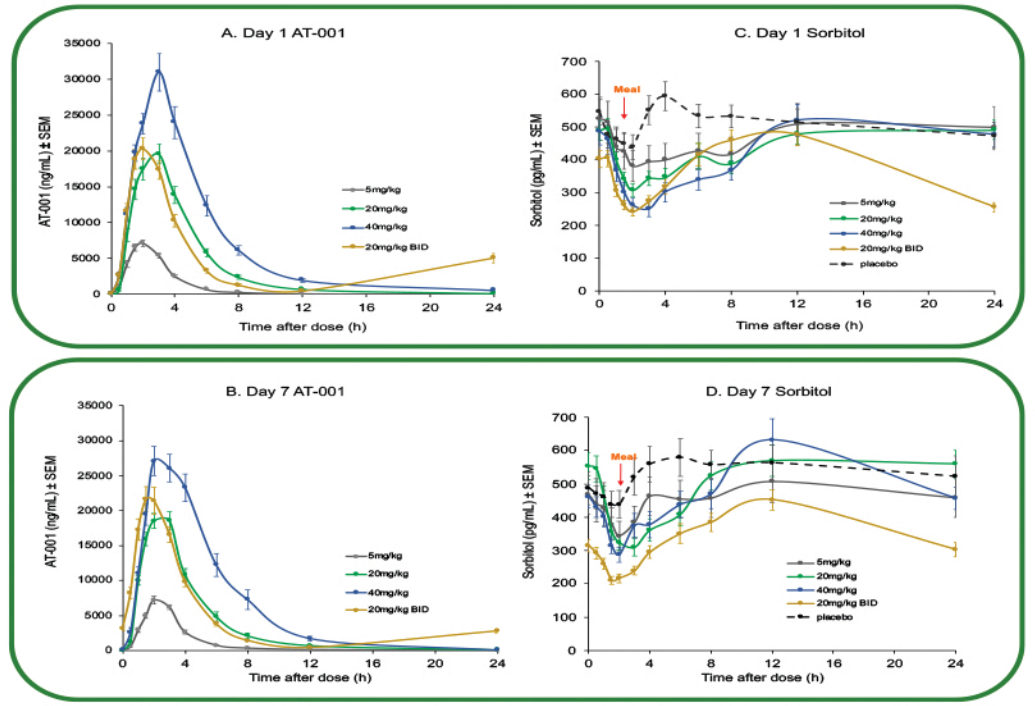
A. Day 1 AT-001
C. Day 1 Sorbitol
B. Day 7 AT-001
D. Day 7 Sorbitol
AT-001 (ng/mL) ± SEM
Sorbitol (pg/mL) ± SEM
Meal
Time after dose (h)
5mg/kg
20mg/kg
40mg/kg
20mg/kg BID
placebo

Sorbitol Change From Baseline

FIGURE 5. Whole blood sorbitol percent change from baseline at the time of maximum AT-001 exposure (Cmax)

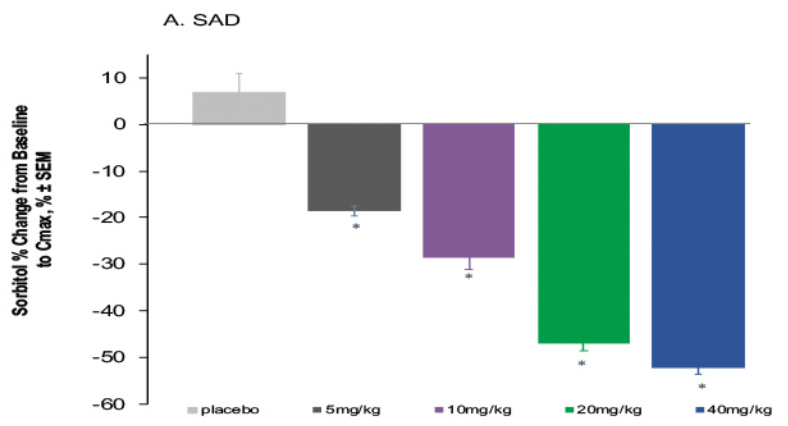
A. SAD
Sorbitol % Change from Baseline to Cmax, % ± SEM
placebo
5mg/kg
10mg/kg
20mg/kg
40mg/kg

Sorbitol Change From Baseline (cont'd)

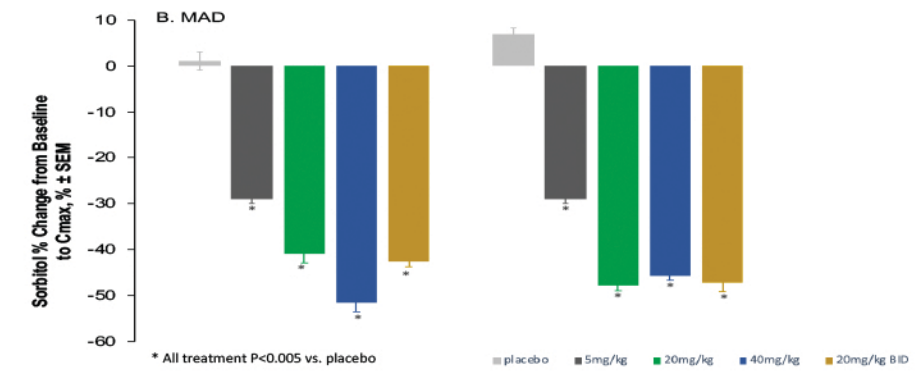
B. MAD
Sorbitol % Change from Baseline to Cmax, % ± SEM
* All treatment P<0.005 vs. placebo
placebo
5mg/kg
20mg/kg
40mg/kg
20mg/kg BID

PK/PD Summary

- Median Tmax was between 1.75 to 3 hrs in both SAD and MAD and T1/2 ranged between 1.74-3.38 hrs across all SAD and MAD cohorts
- Maximal systemic exposure determined from individual concentration-time data (Cmax) and the AUC from time time-zero to infinity (AUC0-inf) increased in a dose proportional manner in SAD and MAD
- There was no evidence of accumulation over 7d in MAD
- PD effects on sorbitol reduction lasted 10-12 hours post dose, demonstrating extended effect of the drug beyond the half-life of the drug; suggested to be due to reversible covalent binding of AT-001 within the active site of the enzyme

Conclusion

- AT-001 significantly reduced cardiac damage in a relevant mouse model of DbCM
- AT-001 improved selectivity and affinity for AR has resulted in potent AR inhibition within a favorable safe dosing range
- In humans, single and multiple ascending doses were well-tolerated with no safety concerns identified
- Proof of biological activity was obtained in all patients dosed
- These findings support further investigation of the therapeutic potential of AT-001 in subjects with Diabetic Cardiomyopathy

References

1. Dandamundi S, et al. J Card Fail. 2014;20(5):304-9
2. Miki T, et al. Hear Failure Rev 2013;18:149-166
3. Parim B, et al Heart Failure Rev 2019:24:279-299
4. Grewal AS, et al. Min Rev Med Chem 2016; 16:120-62
5. Ramasamy et al. Circ Res2010 ;106:1449-58
6. Li Q, et al Cardiovasc Diabetol. 2008;7:33

DISCLOSURES: RP, PR, SS, FL are employees of and stockholders in Applied Therapeutics Inc.

23rd ANNUAL SCIENTIFIC MEETING OF THE HEART FAILURE SOCIETY OF AMERICA, PHILADELPHIA, PA | SEPTEMBER 13-16TH, 2019

APPLIED THERAPEUTICS