A Novel Investigational Treatment in Preclinical Development for Galactosemia

Shauna Rasmussen, PhD[1], Jenna Daenzer, PhD[1], Martine Williams, BS[1], Jessica MacWilliams[1], Shoshana Shendelman, PhD[2], Riccardo Perfetti, MD, PhD[2], and Judith Fridovich-Keil, PhD[1]
1. Emory University School of Medicine, 2. Applied Therapeutics Inc.

INTRODUCTION

- Galactosemia is a potentially life-threatening autosomal recessive metabolic disorder, affecting between 1/30,000-1/60,000 live births, that leads to neonatal, developmental and adult changes in various tissues presumably resulting from the abnormal accumulation of toxic metabolites in the central nervous system, the lens of the eye, liver, kidney and other tissues
- Affected infants are born seemingly healthy, but then experience a rapid and devastating decline following exposure to breast milk or milk formula, which contain large quantities of lactose, which is metabolized to galactose
- Acute symptoms can progress in a matter of days from jaundice, vomiting, and diarrhea to failure to thrive, hepatomegaly, and *E. coli* sepsis. Without rapid dietary restriction of galactose, affected infants often die in the neonatal period
- Despite dietary restriction of galactose, patients with galactosemia can experience severe phenotypes which include cognitive disability, speech defects, tremor, ovarian insufficiency, cataracts, and other adverse outcomes
- Enzyme mutations in the metabolic pathway of galactose utilization are responsible for galactosemia. The resulting deficiency in galactose metabolism leads to an excess activation of the aldose reductase pathway with an overproduction of galactitol, which then accumulates in various tissues (***Figure 1***)
- AT-007 is an Aldose Reductase Inhibitor that blocks the production of galactitol. In disease-specific animal models, inhibition of galactitol has been shown to modify the natural course of the disease

Figure 1. AT-007 Inhibition of Aldose Reductase Reduces Galactitol Formation

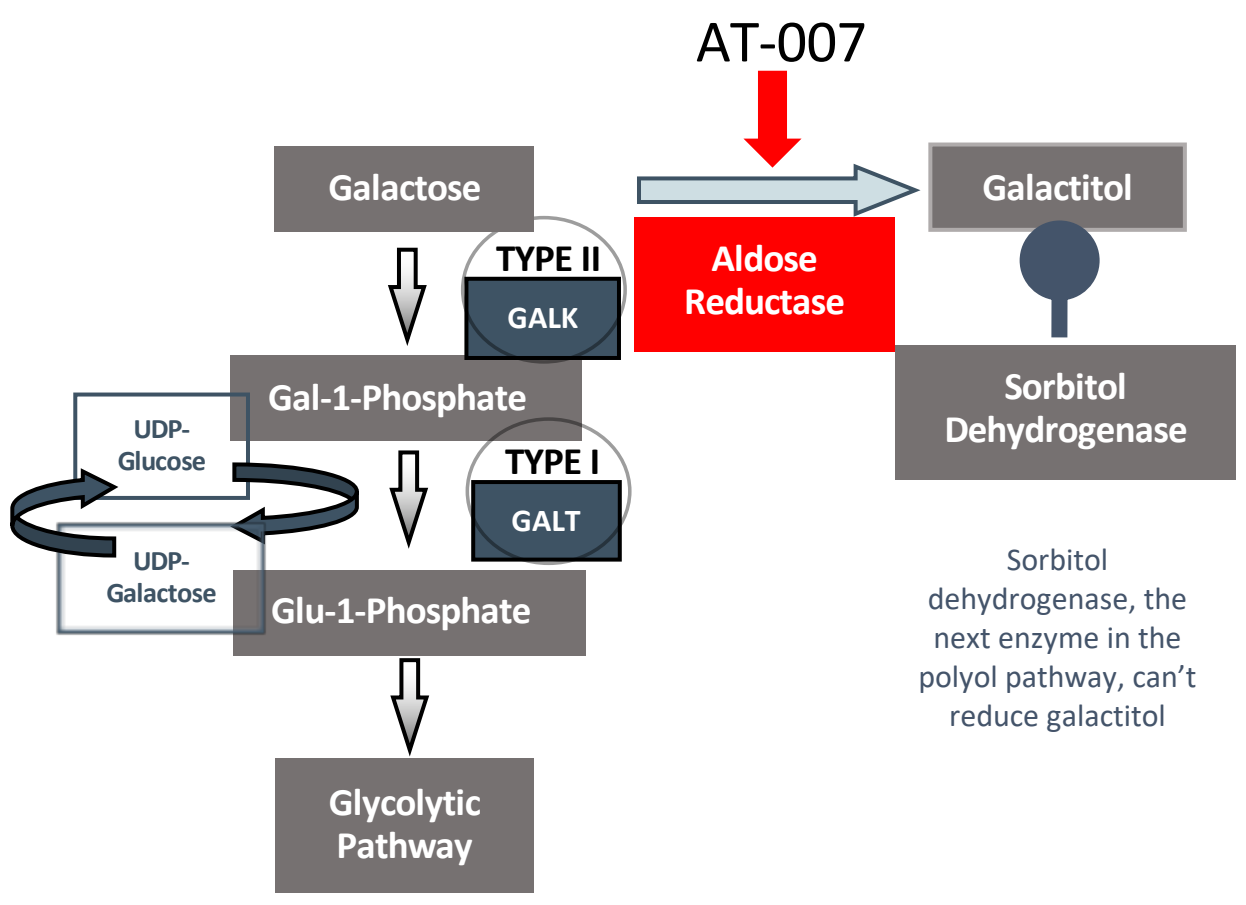
AT-007
Galactose
Aldose Reductase
Galactitol
TYPE II
GALK
Gal-1-Phosphate
Sorbitol Dehydrogenase
UDP-Glucose
UDP-Galactose
TYPE I
GALT
Glu-1-Phosphate
Glycolytic Pathway
Sorbitol dehydrogenase, the next enzyme in the polyol pathway, can't reduce galactitol

STUDY OBJECTIVES

- The aim of this study was to assess the impact of treatment with AT-007, an Aldose Reductase Inhibitor, on formation of cataracts and metabolite levels (galactose & galactitol) in liver, brain and plasma in an animal model of galactosemia (GALT null rat)

METHODS

- *Genetic model:* GALT null rats were homozygous for a frameshift (M3) mutation in the rat *GALT* gene. Wild-Type (WT, +/+) and heterozygous rats (+/M3) exhibited high levels of GALT activity, whereas M3 homozygotes (M3/M3) did not express detectable GALT activity and showed aberrantly high levels of galactose, galactitol, and Gal1P in liver and brain
- Pup *GALT* genotypes (M3/M3 versus M3/+ versus +/+) were determined by Transnetyx using DNA isolated from tail snips collected from all pups at between 5-8 days after birth. Genotypes were corroborated in representative pups by GALT enzyme activity measured in liver samples harvested after euthanasia. In all samples tested, the GALT activity observed was consistent with the anticipated *GALT* genotype
- *Treatment*: Animal treatment began on the day after birth (neonatal Day 1), with the day of birth being designated Day 0. Animals were treated with either placebo (vehicle) or drug (AT-007) for 10 or 22 consecutive days and then were euthanized on Day 10 or Day 22
- Vehicle used to administer the drug to newborn pups was Gerber Good Start Birth-12 Months soy formula, prepared according to the manufacturer's instructions. Drug (AT-007) powder, suspended in freshly prepared Gerber formula, was administered orally once daily at 1mg AT-007/gram pup weight for each dose. Placebo and drug were fed to pups by hand using a syringe and nipple + tubing
- *Galactose and metabolites:* Galactose, Gal1P and galactitol metabolites were resolved and quantified via HPLC in samples of liver, brain and plasma from euthanized pups. Tissue samples were assayed as previously described (Ross et al. 2004 Mol Gen Metab 83:103-116 and Daenzer et al. 2016 Dis Models and Mech 9:1375-1382)
- *Tracking/ quantifying cataracts*: Cataracts were scored visually by a blinded rater on a 4-point scale, with zero being no cataracts present (absent), 1 being mild, 2 being moderate and 3 being severe cataracts present. Severity of cataracts was scored based on both size and opacity of the eye lens. In addition, animals exposed to the study drug or placebo were photographed via digital camera, and the opacity of the lens quantified via custom software

RESULTS

- Treatment with AT-007 led to significant reductions in galactitol in the GALT null weanling rat in all tested tissues, including blood, brain and liver (***Figure 2***)
- AT-007 treatment did not have a significant effect on galactose or Gal1P in this model (***Figure 3***)
- AT-007 completely prevented cataract formation in the GALT null rat, which uniformly developed severe cataracts when untreated (***Figures 4-6***)

Figure 2. Galactitol Levels (quantified by HPLC) at Day 22

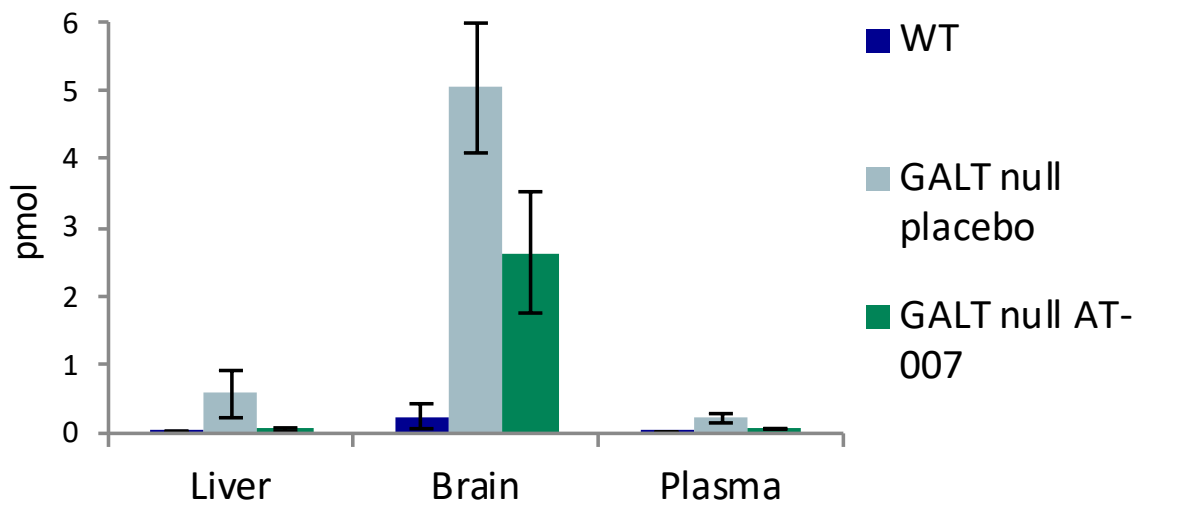
6
5
4
3
2
1
0
pmol
Liver
Brain
Plasma
WT
GALT null placebo
GALT null AT-007

Figure 3. Galactose and Gal1P Levels (quantified by HPLC) at Day 22

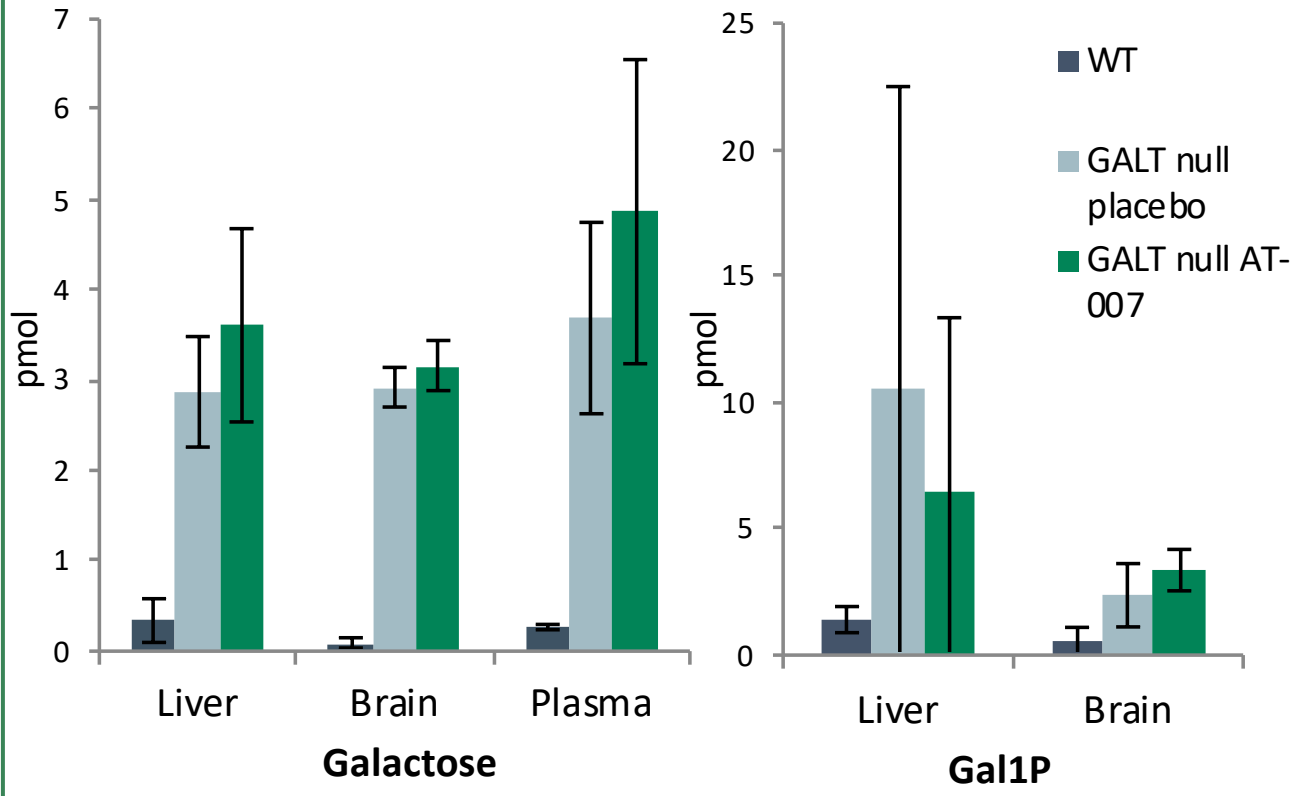
7
6
5
4
3
2
1
0
pmol
Liver
Brain
Plasma
Galactose
25
20
15
10
5
0
pmol
Liver
Brain
Gal1P
WT
GALT null placebo
GALT null AT-007

Figure 4. Digital Ophthalmic Quantitation Of Cataract Formation at Day 22

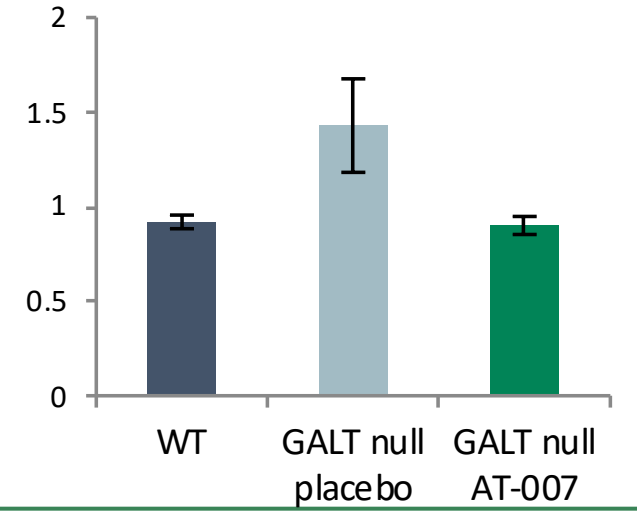
2
1.5
1
0.5
0
WT
GALT null placebo
GALT null AT-007

Figure 5. Cataract Severity Score (0-3) at Day 22

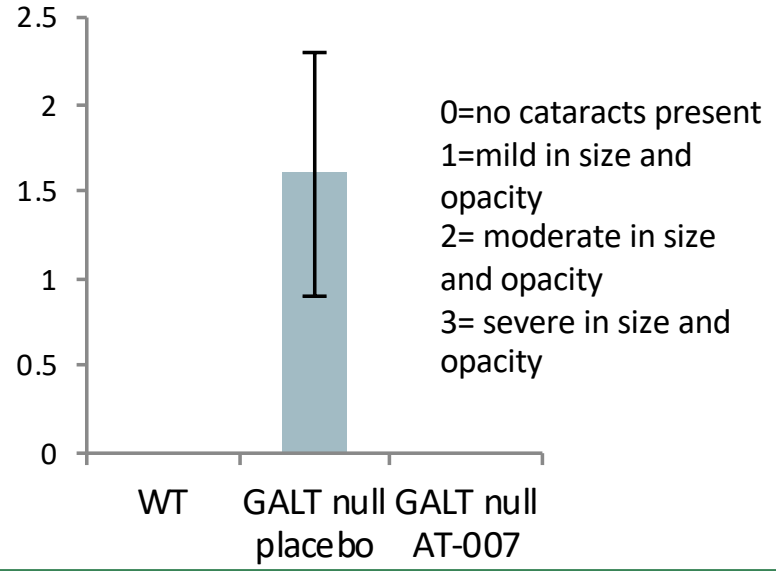
2.5
2
1.5
1
0.5
0
WT
GALT null placebo
GALT null AT-007
0=no cataracts present
1=mild in size and opacity
2= moderate in size and opacity
3= severe in size and opacity

Figure 6. Cataract Formation at Day 22

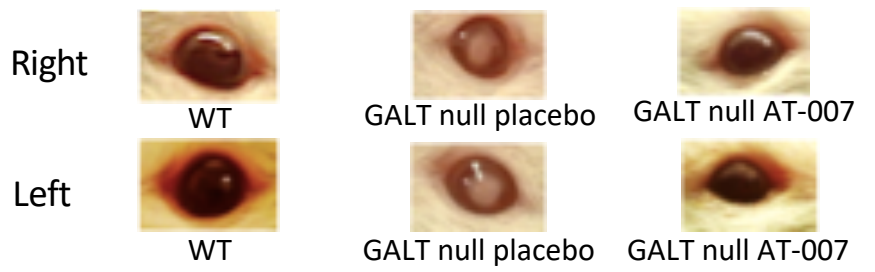
Right
Left
WT
GALT null placebo
GALT null AT-007
WT
GALT null placebo
GALT null AT-007

CONCLUSIONS

- Galactosemia is an autosomal recessive disease that can be fatal in newborns and is associated with long-term complications, including presenile cataracts, CNS-related abnormalities, low bone density and primary ovarian insufficiency
- While dietary restriction of lactose and galactose limits exogenous intake of galactose and prevents fatalities in infancy, it does not address the consequences of endogenous production of galactose, which significantly impacts development in children, and quality of life in adulthood
- AT-007 (a novel ARI) effectively reduced galactitol levels in the GALT null rat model of classic galactosemia across relevant tissues, and prevented development of pre-senile cataracts in this model
- This study confirms the hypothesis that galactitol is responsible for cataract formation in GALT deficiency, and treatment with an ARI can prevent formation of cataracts. Studies are ongoing to determine whether other long-term complications, such as CNS deficiencies, may be caused by elevated galactitol, and if treatment with AT-007 can prevent other long-term complications. AT-007 represents a potentially important new strategy for prevention and treatment of galactosemia complications